The investigational drug ZEN-3694, a novel BET-bromodomain inhibitor, inhibits multiple tumor immune escape mechanisms and has the potential to combine with immunotherapies

Sarah Attwell[1], Karen Norek[1], Ravi Jahagirdar[1], Cyrus Calosing[1], Sanjay Lakhotia[2], Eric Campeau[1], Henrik C. Hansen[1]
[1]Zenith Epigenetics, Suite 300, 4820 Richard Road SW, Calgary AB, Canada and [2]Suite 4010, 44 Montgomery St. San Francisco CA, USA,

Abstract

ZEN-3694 is an orally bioavailable, potent inhibitor of both bromodomains of the Bromodomain and Extra-Terminal domain (BET) proteins. *In vitro*, ZEN-3694 has demonstrated strong activity against cell lines representing a broad range of solid tumor and hematological malignancies with submicromolar potency, and shows *in vivo* activity in several xenograft models, including prostate, breast, colon and AML. ZEN-3694 is currently in phase 1 clinical trials for metastatic castration resistant prostate cancer patients who have progressed on enzalutamide and/or abiraterone.

Immunotherapies targeting the PD1/PD-L1 axis have shown remarkable durable efficacy for many cancers, but still, the majority of patients do not respond to these therapies alone, and therefore there is a need to identify combination agents which will increase the response rate.

Here we show that ZEN-3694 targets many pathways which suppress the anti-tumor immune response. In solid tumor cell lines representing a variety of malignancies, ZEN-3694 downregulates the checkpoints B7H3 and PD-L1, and upregulates the MICA antigen. In activated CD8+ T cells, ZEN-3694 targets multiple checkpoint receptors known to be involved in tumor escape. ZEN-3694 also inhibits the differentiation and function of Regulatory T cells (Tregs), and strongly inhibits the suppressive cytokines/chemokines IL-10 and CCL2. ZEN-3694 also targets several recently identified markers of intrinsic PD-1 resistance, and inhibits the angiogenic factor VegF. Effects on these various markers of immunomodulation are being confirmed in our current phase I study.

Immunomodulatory effects were also measured *in vivo.* In an MC-38 colon cancer syngeneic xenograft model, the addition of ZEN-3694 increases the efficacy of anti-PD1 in tumor growth inhibition. The ZEN-3694 treated mice showed a significant increase in IFNg+ CD8 T cells in the draining lymph nodes, as well as an increase in CD8+ tumor infiltrating lymphocytes (TILS) in the tumor. Analysis of the tumors showed a decrease in markers of myeloid suppressive cells.

Taken together these data suggest that ZEN-3694 targets several mechanisms of resistance to PD1 therapy, and has the potential to synergize with a variety of cancer immunotherapies.

Background

The Bromodomain and Extra-Terminal domain (BET) family of proteins BRD2, BRD3, BRD4, and BRDT are epigenetic readers that bind via their tandem bromodomains (BD1 & BD2) to acetylated lysines in histones and promote gene transcription. Tumor type specific super-enhancers associated with key oncogenes involved in tumor pathogenesis have been identified in hematological as well as solid tumor malignancies[1,2]. Inhibition of BET proteins results in their displacement from super-enhancers leading to down regulation of key oncogenic programs, including members of the MYC and BCL-2 families[1]. Additionally, BET inhibitors (BETi) target pathways involved in metastasis, such as the NF-kB and Wnt5a pathways[3,4]. BET inhibitors have been demonstrated to inhibit proliferation and suppress tumorigenicity in numerous solid and hematological malignancies.

Although PD1 antibodies have shown remarkable and durable efficacy in a portion of cancers, a variety of immune mechanisms contribute to both innate and acquired resistance in the majority of patients. These include upregulation of alternate tumor and T cell checkpoint receptors, recruitment of suppressive cells which dampen the T cell response, and tumor mechanisms to decrease immune recognition. Here we show that BET inhibitors have an additional unique mechanism of action: they inhibit multiple complementary mechanisms of tumor immune escape, which suggest that they could synergize with immunotherapies.

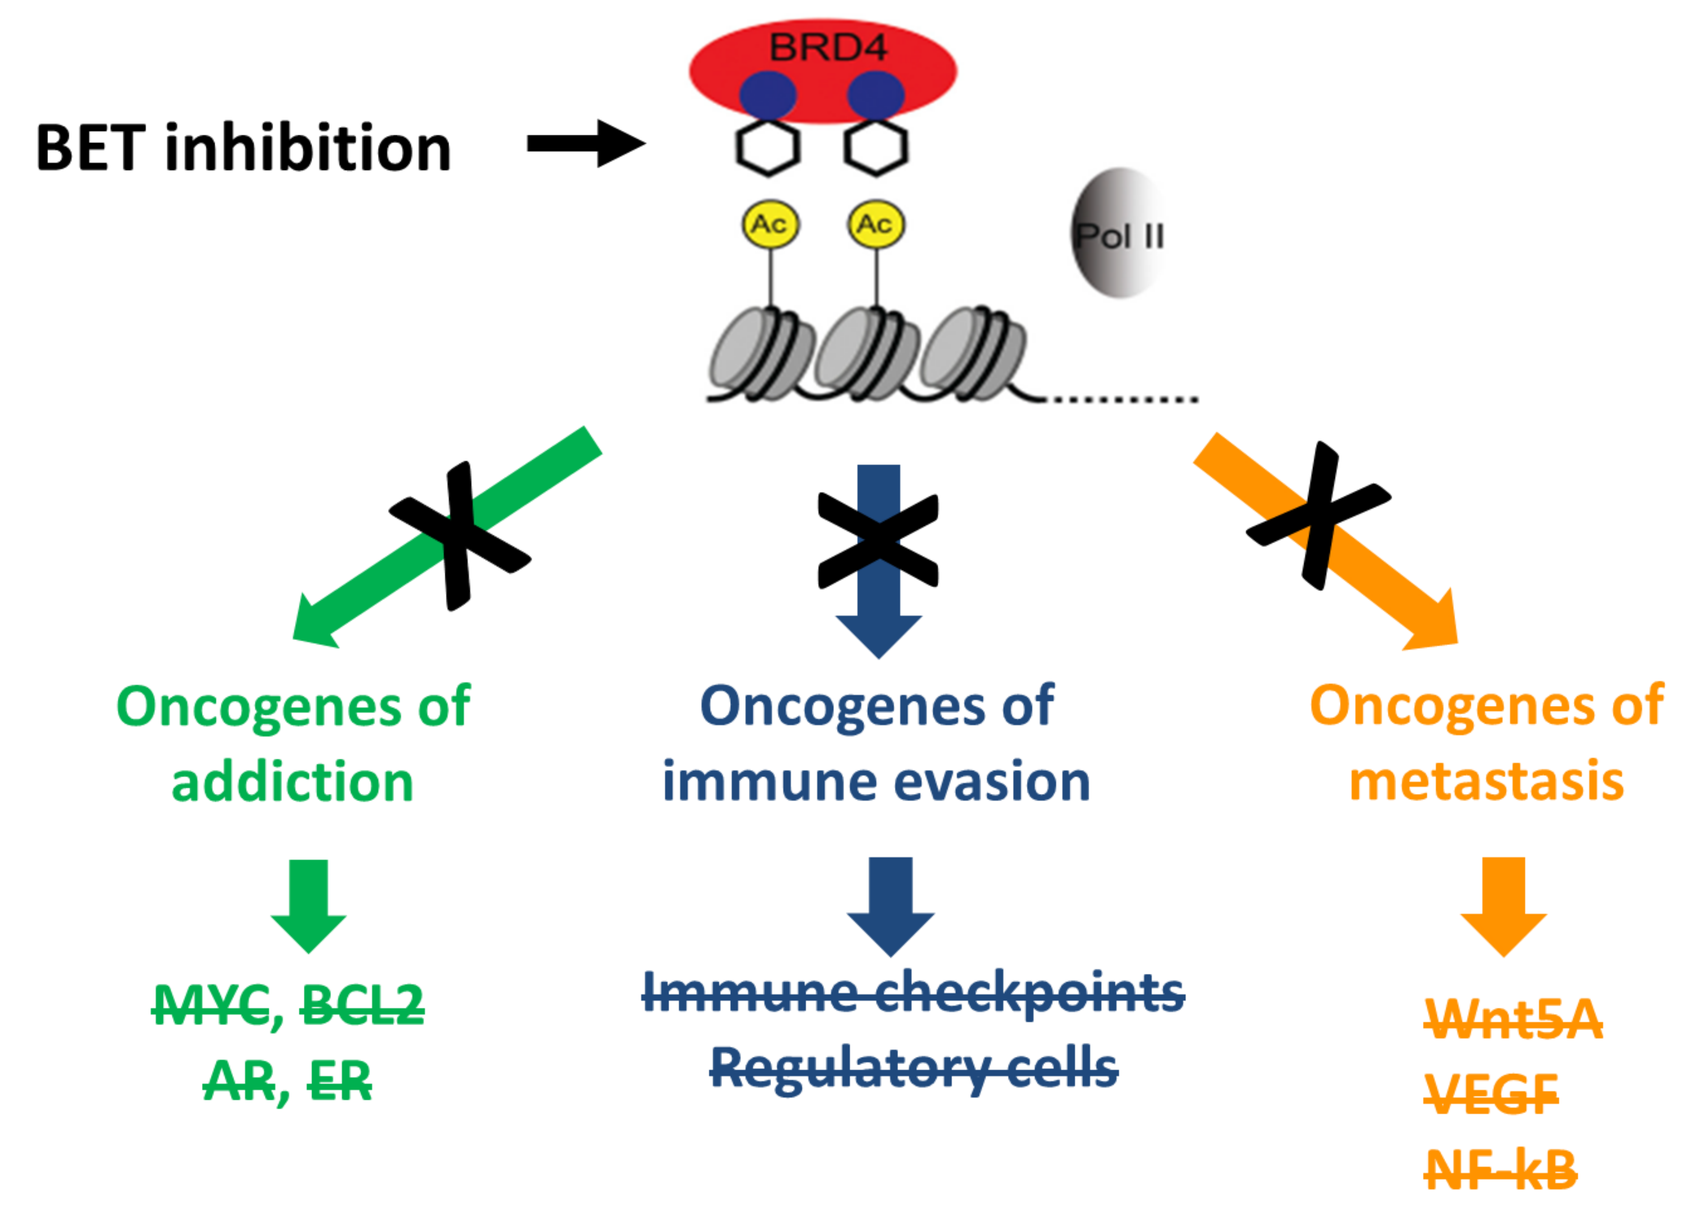
BET inhibition
BRD4
Ac
Pol II
Oncogenes of addiction
~~MYC, BCL2~~
~~AR, ER~~
Oncogenes of immune evasion
~~Immune checkpoints~~
~~Regulatory cells~~
Oncogenes of metastasis
~~Wnt5A~~
~~VEGF~~
~~NF-kB~~

Results

ZEN-3694 targets a unique set of checkpoint receptors, cytokines, and tumor factors

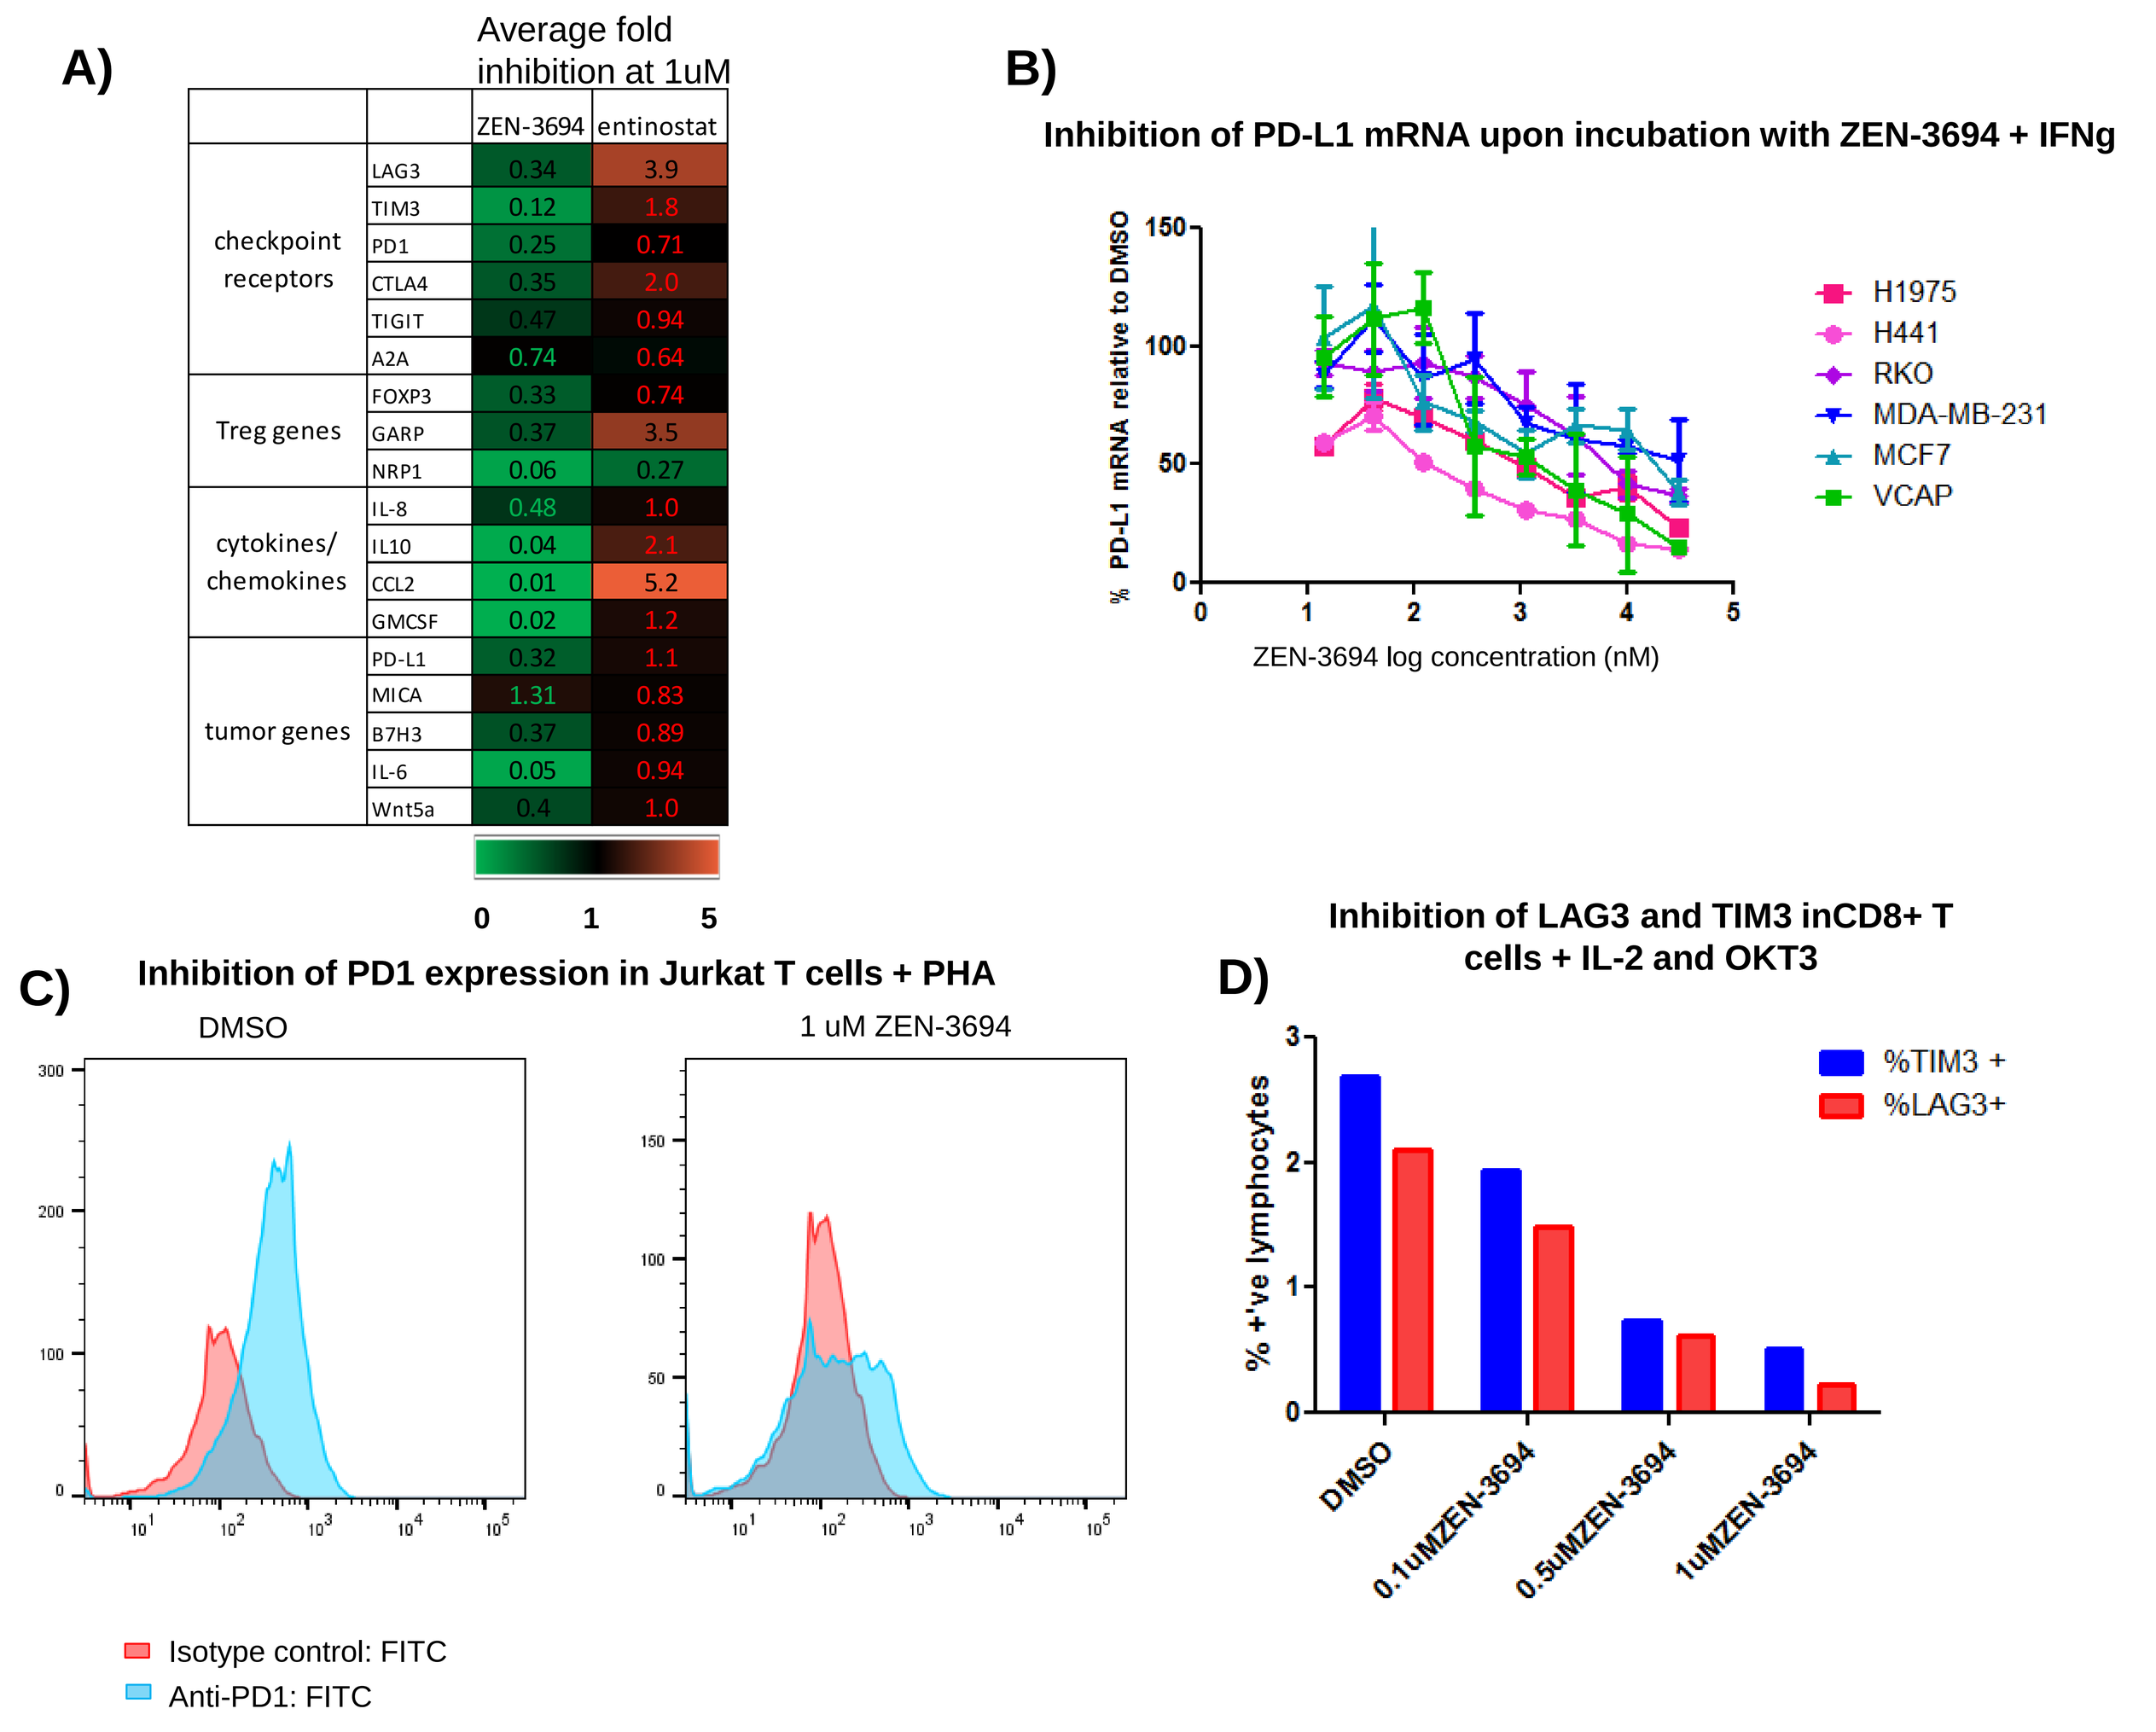
A)
Average fold inhibition at 1uM
ZEN-3694 entinostat
checkpoint receptors
LAG3 0.34 3.9
TIM3 0.12 1.8
PD1 0.25 0.71
CTLA4 0.35 2.0
TIGIT 0.47 0.94
A2A 0.74 0.64
Treg genes
FOXP3 0.33 0.74
GARP 0.37 3.5
NRP1 0.06 0.27
cytokines/ chemokines
IL-8 0.48 1.0
IL10 0.04 2.1
CCL2 0.01 5.2
GMCSF 0.02 1.2
tumor genes
PD-L1 0.32 1.1
MICA 1.31 0.83
B7H3 0.37 0.89
IL-6 0.05 0.94
Wnt5a 0.4 1.0
0 1 5
B)
Inhibition of PD-L1 mRNA upon incubation with ZEN-3694 + IFNg
% PD-L1 mRNA relative to DMSO
ZEN-3694 log concentration (nM)
H1975
H441
RKO
MDA-MB-231
MCF7
VCAP
C)
Inhibition of PD1 expression in Jurkat T cells + PHA
DMSO
1 uM ZEN-3694
Isotype control: FITC
Anti-PD1: FITC
D)
Inhibition of LAG3 and TIM3 inCD8+ T cells + IL-2 and OKT3
% +'ve lymphocytes
%TIM3 +
%LAG3+
DMSO
0.1uMZEN-3694
0.5uMZEN-3694
1uMZEN-3694

Figure 2. ZEN-3694 inhibits mRNA expression of checkpoint receptors, cytokines, and tumor antigens, and the cell surface expression of the checkpoints PD-1, LAG3, and TIM3. A) BET inhibitors are unique in their capacity to inhibit several important markers involved in tumor immune escape compared to entinostat (shown) and other epigenetic inhibitors (not shown). Gene expression was measured in PBMCs, or A549 lung cancer cells. B) ZEN-3694 inhibits PD-L1 mRNA expression in a variety of solid tumor cancer cell lines. C) Treatment of Jurkat T cells with ZEN-3694 reduces the levels of cell surface PD-1 D) Dose-dependent inhibition of cell surface levels of LAG3 and TIM3 checkpoints upon treatment of CD8+T cells with ZEN-3694

ZEN-3694 inhibits Treg differentiation and function

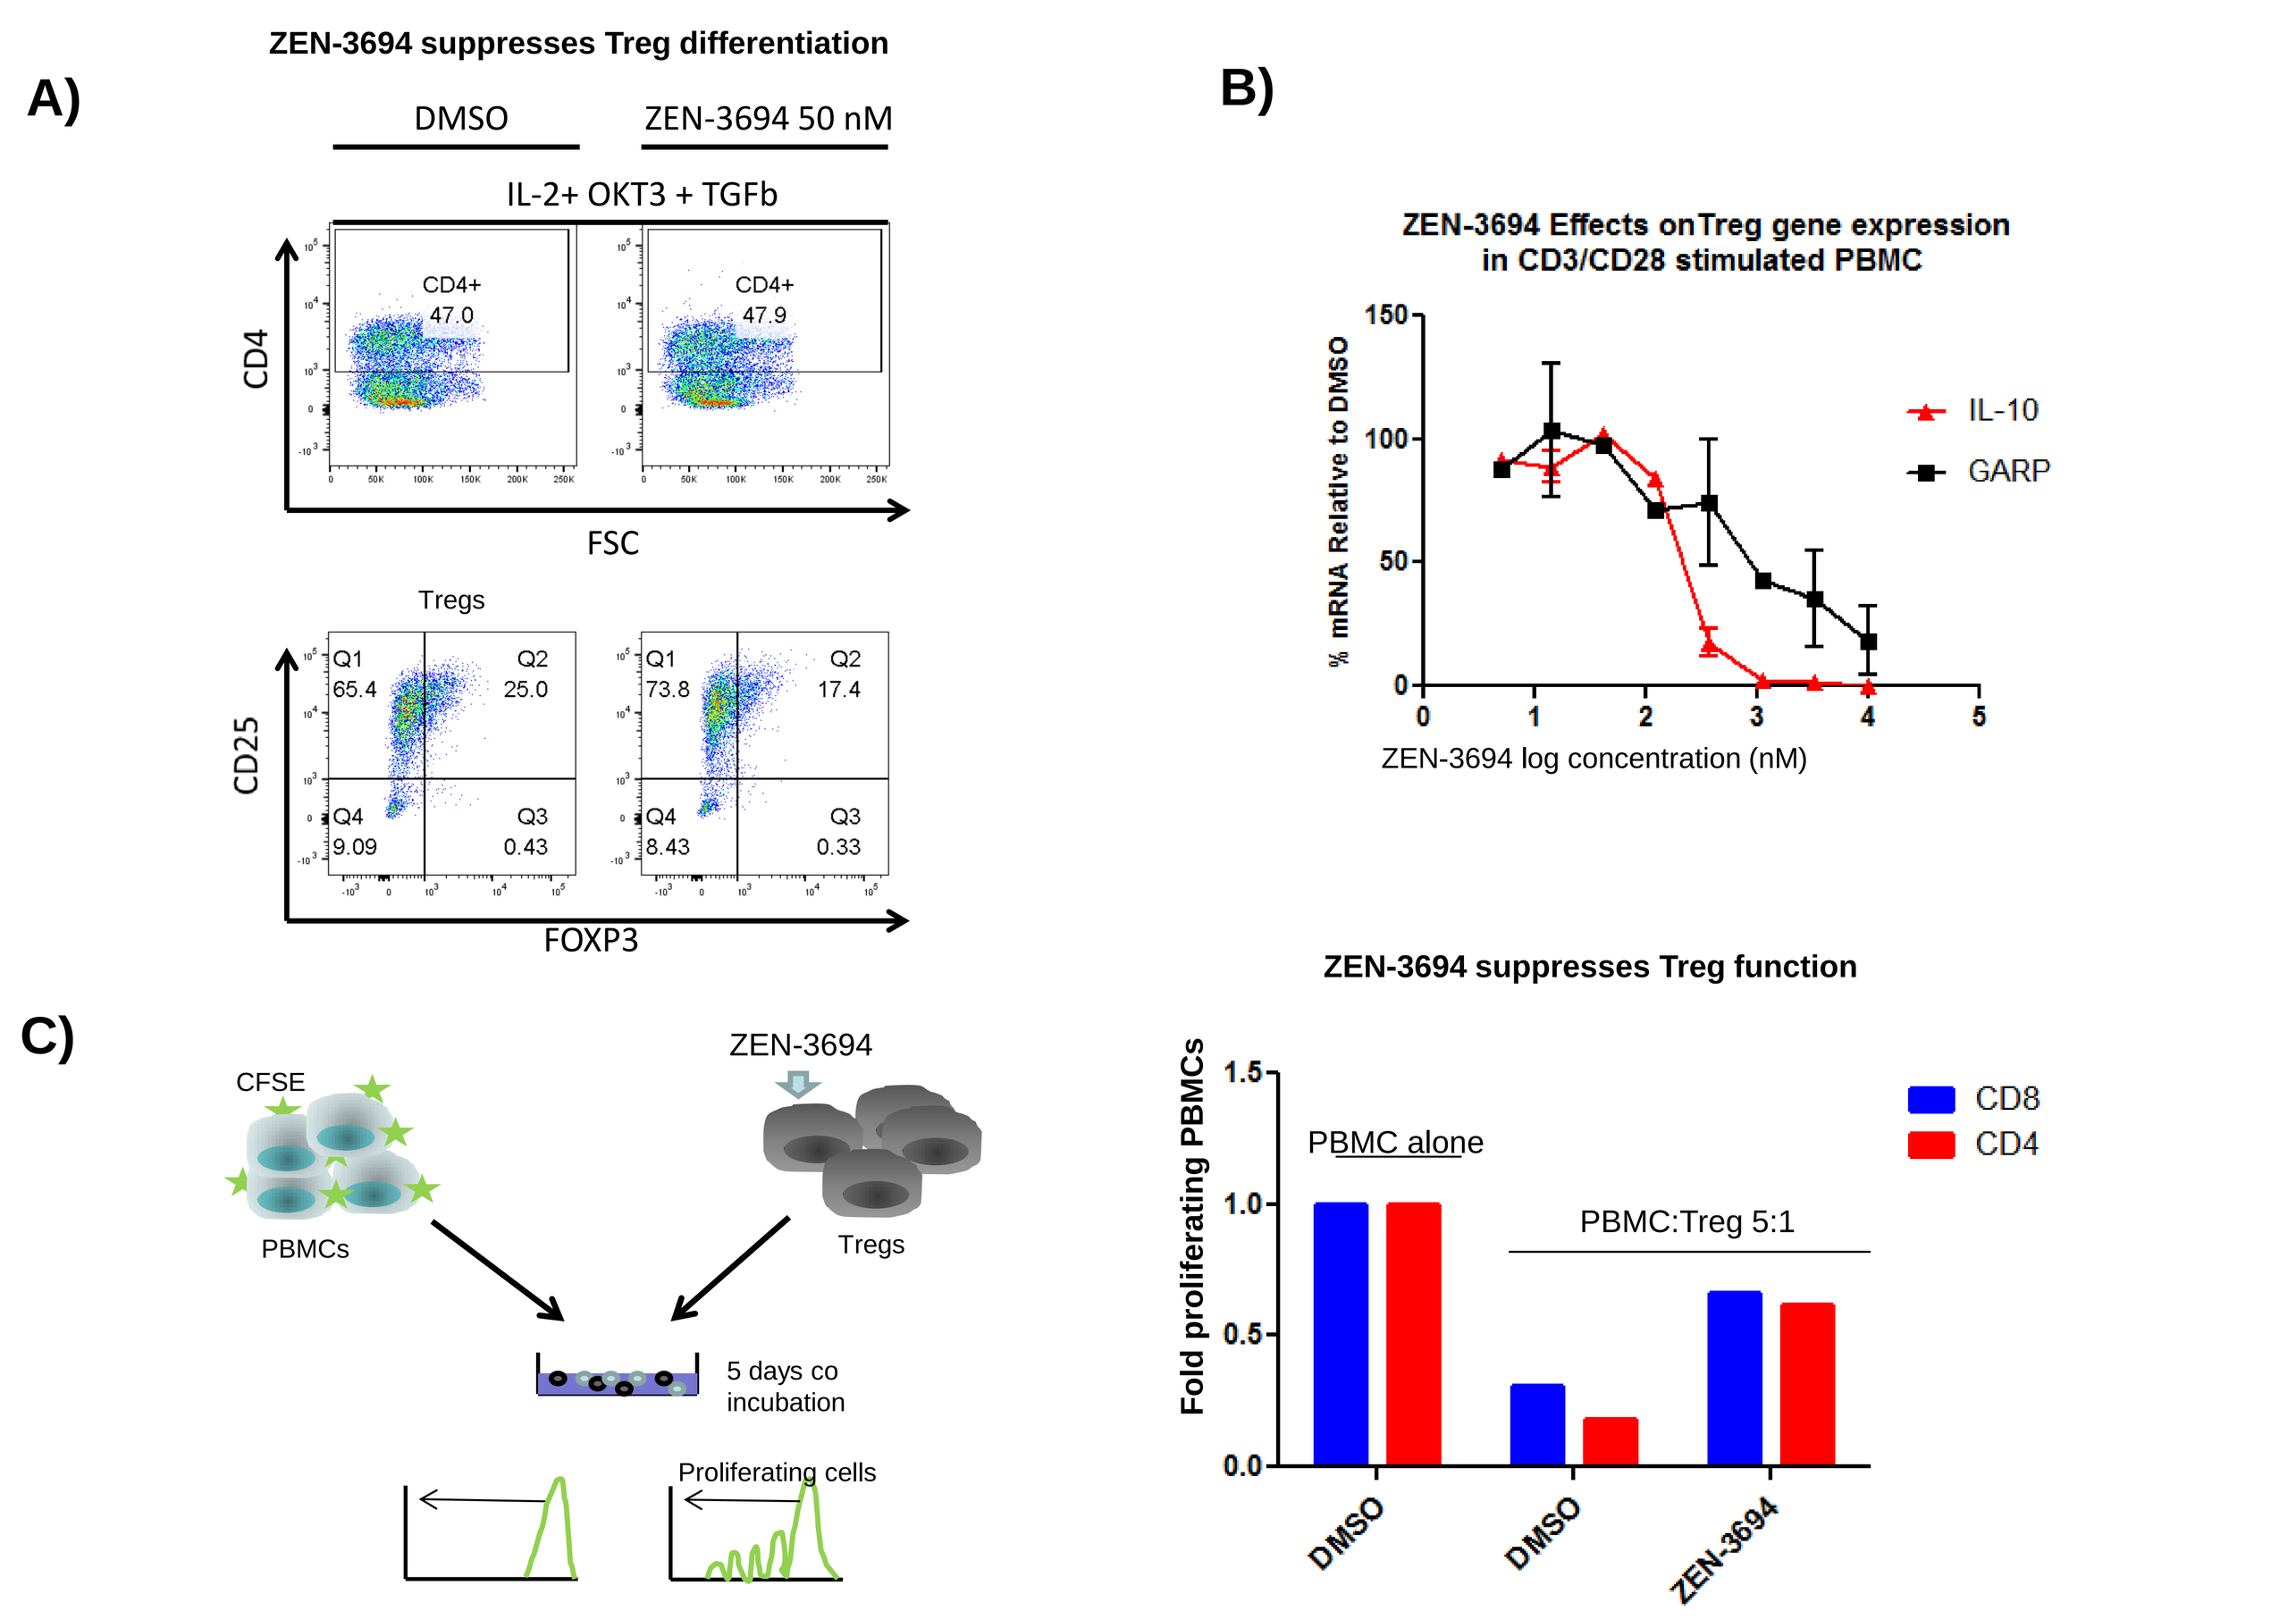
A)
ZEN-3694 suppresses Treg differentiation
DMSO
ZEN-3694 50 nM
IL-2+ OKT3 + TGFb
CD4+ 47.0
CD4+ 47.9
CD4
FSC
Tregs
Q1 65.4
Q2 25.0
Q4 9.09
Q3 0.43
Q1 73.8
Q2 17.4
Q4 8.43
Q3 0.33
CD25
FOXP3
B)
ZEN-3694 Effects onTreg gene expression in CD3/CD28 stimulated PBMC
% mRNA Relative to DMSO
ZEN-3694 log concentration (nM)
IL-10
GARP
C)
CFSE
PBMCs
ZEN-3694
Tregs
5 days co incubation
Proliferating cells
ZEN-3694 suppresses Treg function
Fold proliferating PBMCs
PBMC alone
PBMC:Treg 5:1
CD8
CD4
DMSO
DMSO
ZEN-3694

Figure 3. ZEN-3694 inhibits Treg differentiation, activation, and function. A) Tregs were differentiated from PBMCs. Incubation with ZEN-3694 inhibits the differentiation of Treg (BOTTOM) without affecting the number of CD4+ T cells (TOP) B) ZEN-3694 inhibits the mRNA of two markers of activated Treg cells, IL-10 and GARP. C) LEFT: schematic of experiment for Treg functional assay: Tregs were pre-treated with ZEN-3694, and then incubated with PBMCs stimulated to proliferate. Proliferating PBMCs gradually lose the CFSE label. RIGHT: Pre-incubation of Treg cells with ZEN-3694 decrease their inhibitory potential which results in increase proliferation of PBMCs.

ZEN-3694 increases markers of tumor immunogenicity

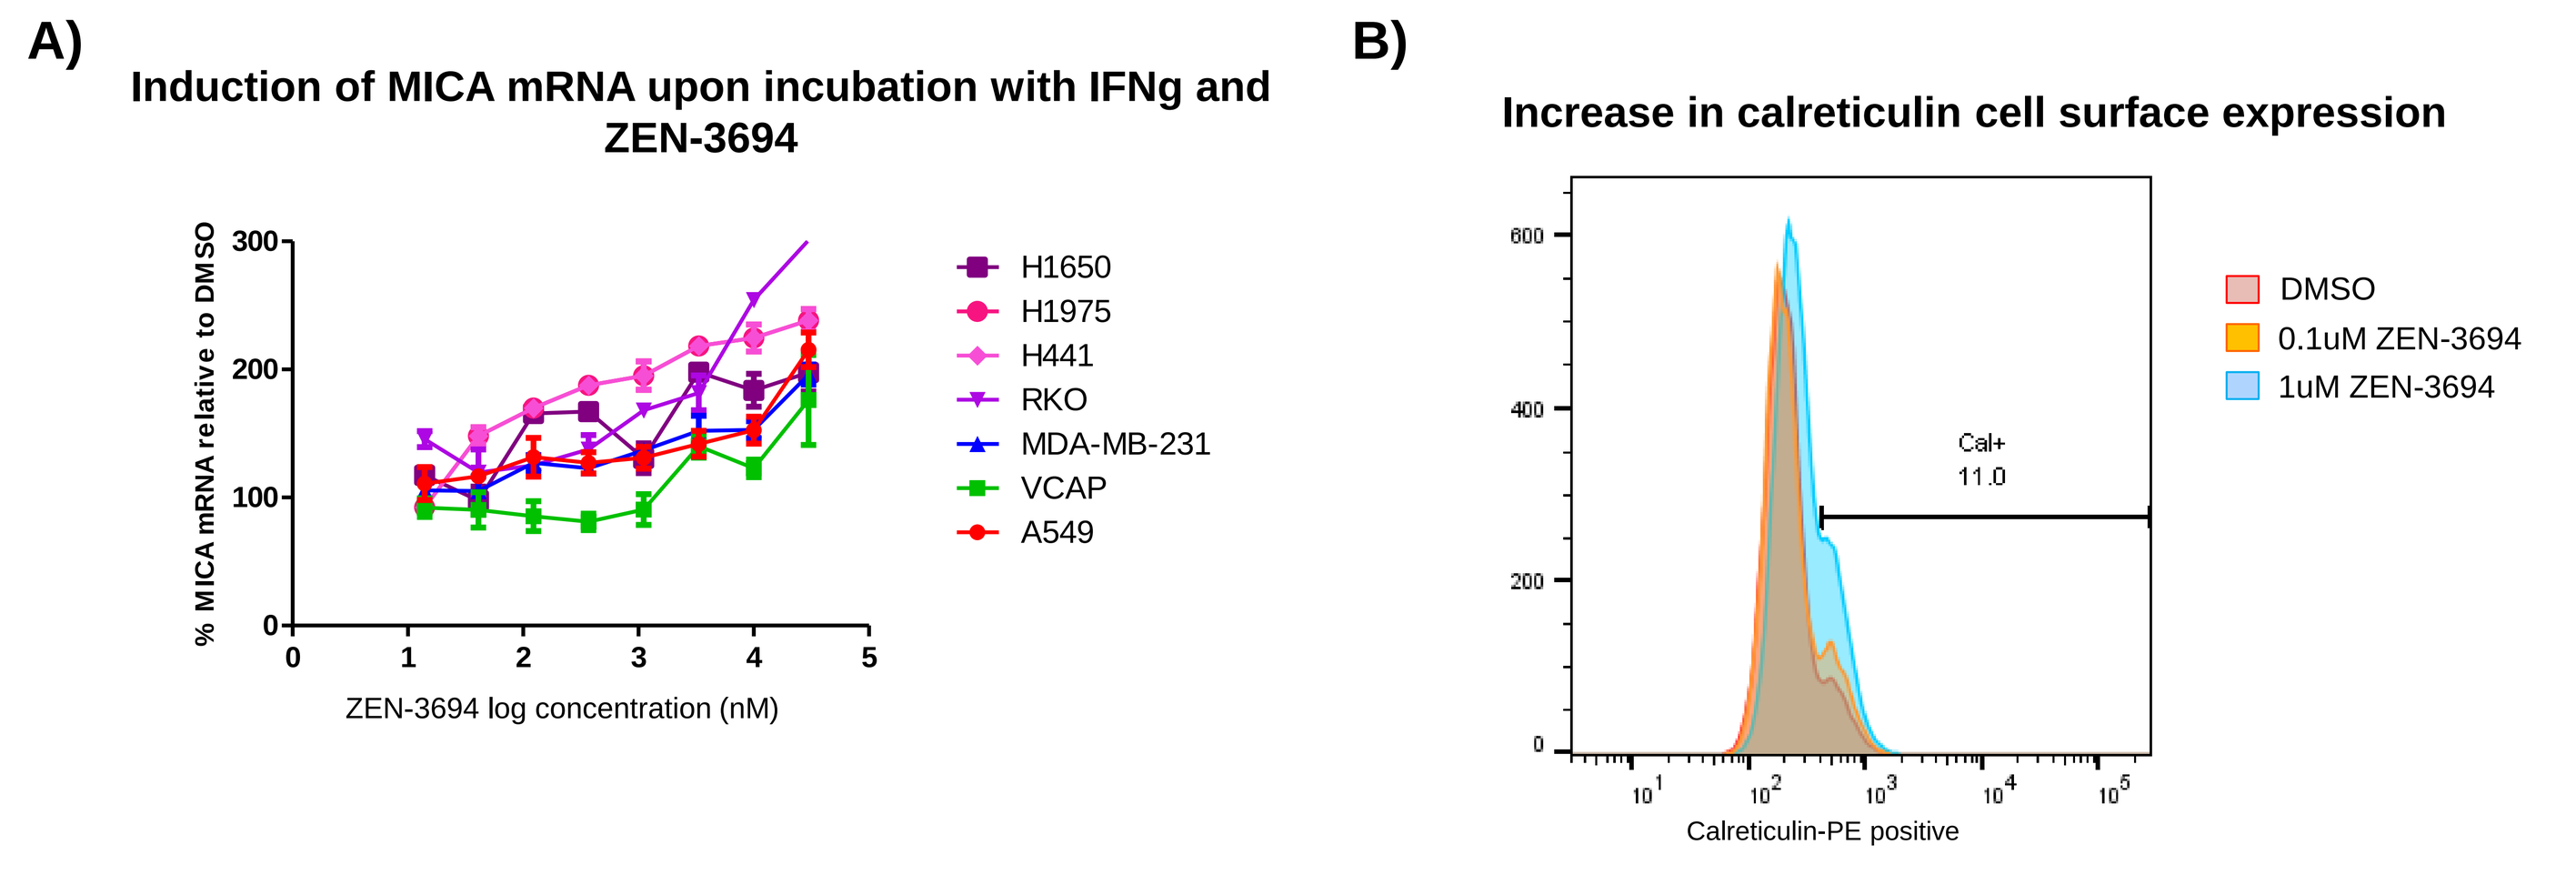
A)
Induction of MICA mRNA upon incubation with IFNg and ZEN-3694
% MICA mRNA relative to DMSO
ZEN-3694 log concentration (nM)
H1650
H1975
H441
RKO
MDA-MB-231
VCAP
A549
B)
Increase in calreticulin cell surface expression
Cal+ 11.0
Calreticulin-PE positive
DMSO
0.1uM ZEN-3694
1uM ZEN-3694

Figure 4. ZEN-3694 induces expression of the NK cell ligand MICA, and the cell surface expression of calreticulin, a phagocytosis marker for dendritic cells. A) ZEN-3694 increases mRNA levels of the major histocompatibility complex class I polypeptide-related sequence A (MICA) in a variety of solid tumor cell lines, without increasing shedding of MICA in the media (not shown). B) MV4-11 cells were incubated for 24 hours with ZEN-3694, which resulted in the increase of cell surface calreticulin, which is associated with increased immunogenicity of tumors, also known as immunogenic cell death.

.

ZEN-3694 targets multiple suppressive factors, and influences T cell activation in MC-38 colon cancer syngeneic mouse models

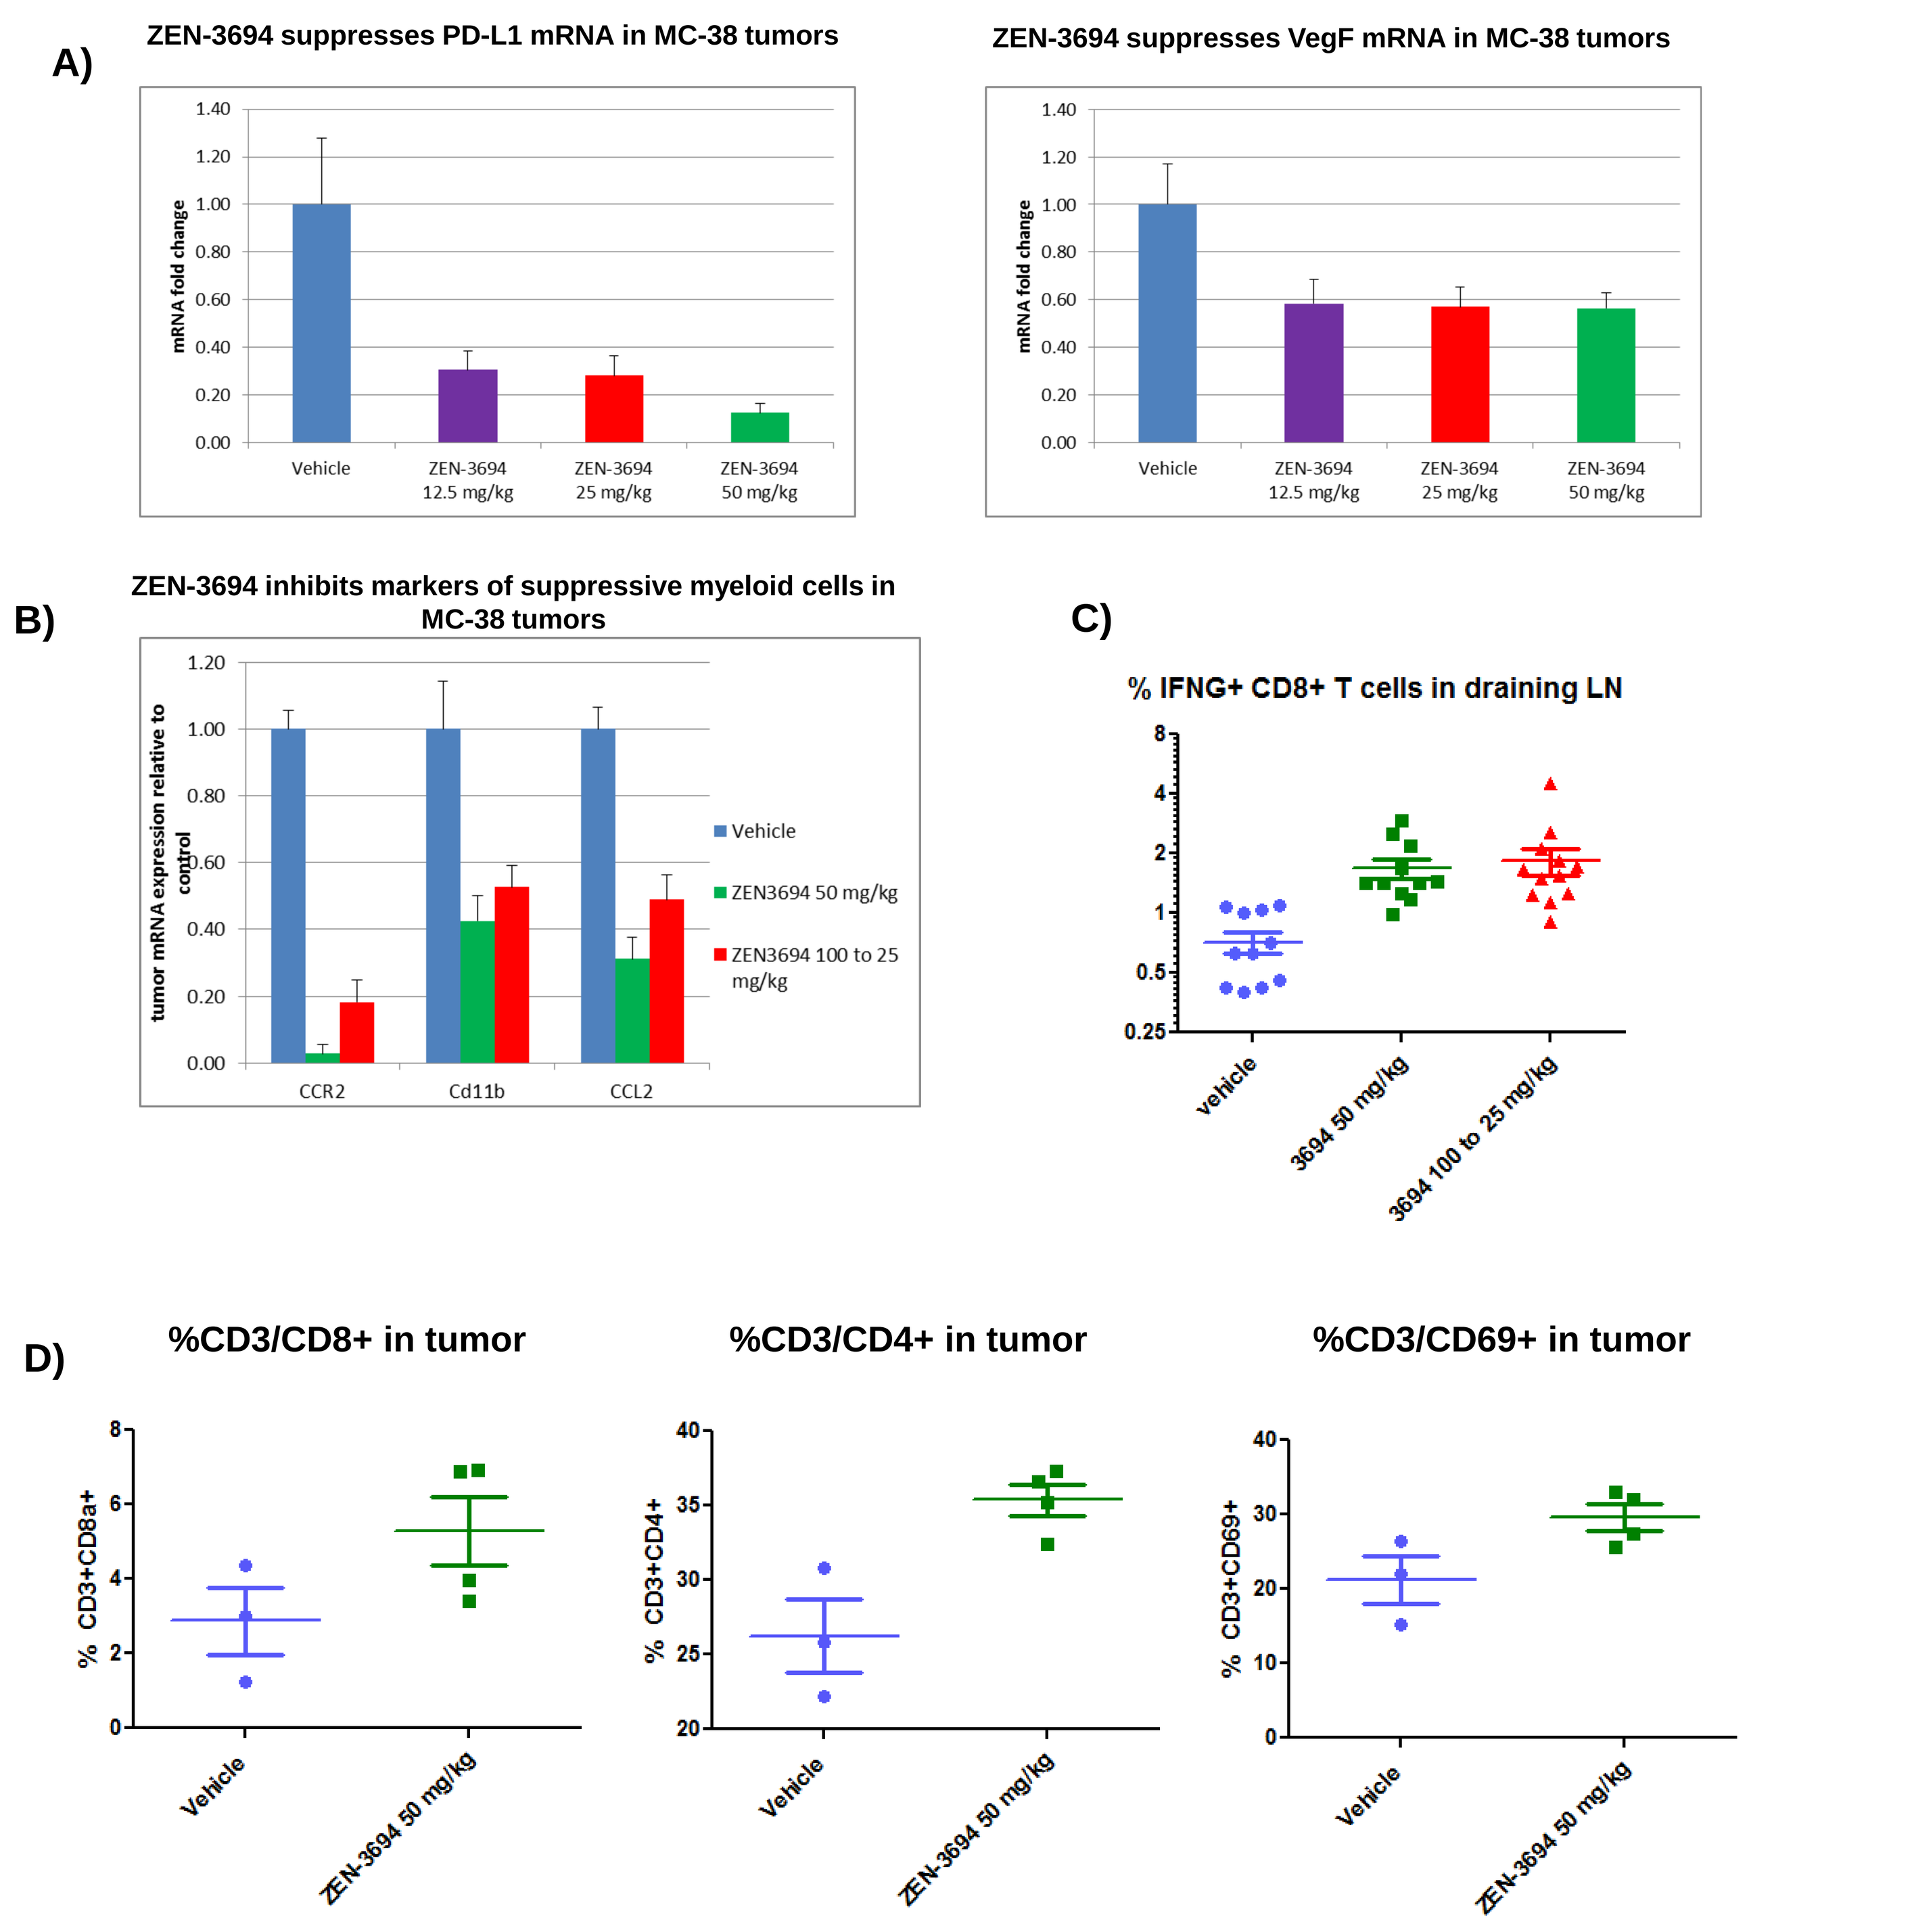
A)
ZEN-3694 suppresses PD-L1 mRNA in MC-38 tumors
ZEN-3694 suppresses VegF mRNA in MC-38 tumors
mRNA fold change
Vehicle
ZEN-3694 12.5 mg/kg
ZEN-3694 25 mg/kg
ZEN-3694 50 mg/kg
B)
ZEN-3694 inhibits markers of suppressive myeloid cells in MC-38 tumors
tumor mRNA expression relative to control
Vehicle
ZEN3694 50 mg/kg
ZEN3694 100 to 25 mg/kg
CCR2
Cd11b
CCL2
C)
% IFNG+ CD8+ T cells in draining LN
vehicle
3694 50 mg/kg
3694 100 to 25 mg/kg
D)
%CD3/CD8+ in tumor
%CD3/CD4+ in tumor
%CD3/CD69+ in tumor
% CD3+CD8a+
% CD3+CD4+
% CD3+CD69+
Vehicle
ZEN-3694 50 mg/kg

Figure 5. ZEN-3694 inhibits expression of PD-L1 mRNA and other immune suppressive markers in MC-38 tumors, and increases T cell activation in draining lymph nodes and tumors A) Treatment of mice with ZEN-3694 results in a decrease in PD-L1 and VEGFA tumor mRNA B) Decrease in CCR2, CD11b, and CCL2 , markers of the suppressive TAM and MDSC response. C) ZEN-3694 induces an increase in activated (IFNG+) CD8+ T cells in the draining lymph node D) ZEN-3694 increases TIL infiltration into the tumor, including CD8, CD4, and total activated (CD69+) T cells

ZEN-3694 is synergistic with anti-PD1 in the MC-38 syngeneic mouse model

A)

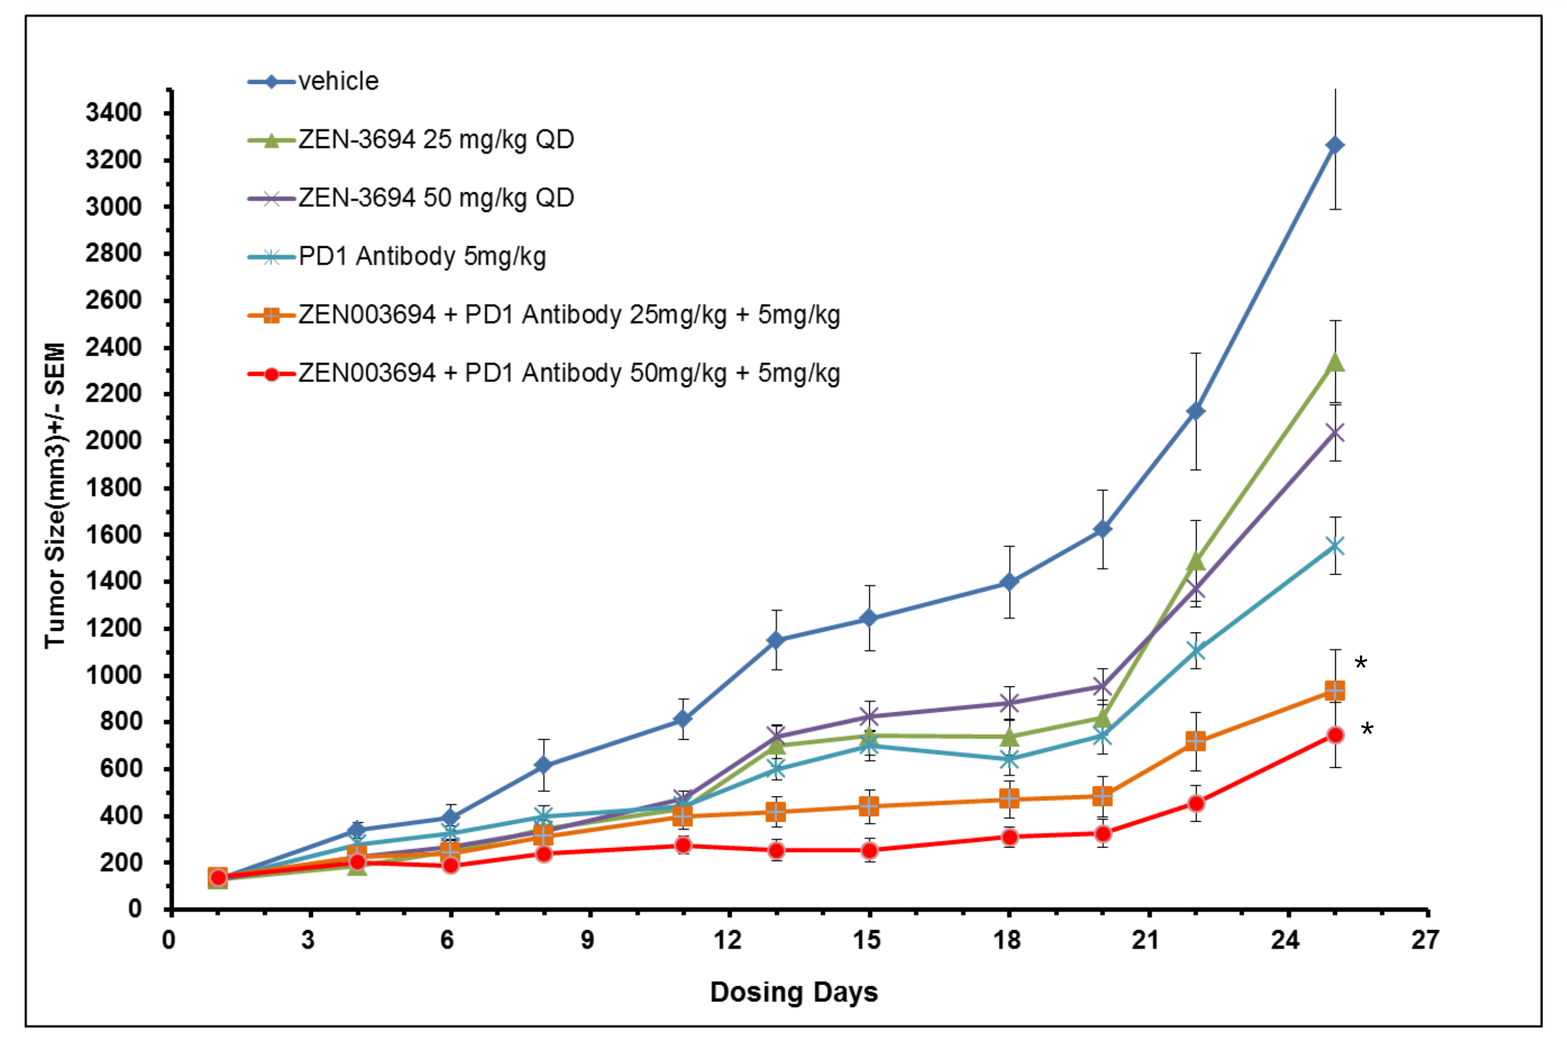
vehicle
ZEN-3694 25 mg/kg QD
ZEN-3694 50 mg/kg QD
PD1 Antibody 5mg/kg
ZEN003694 + PD1 Antibody 25mg/kg + 5mg/kg
ZEN003694 + PD1 Antibody 50mg/kg + 5mg/kg
Tumor Size(mm3)+/- SEM
Dosing Days

*p=<0.01 relative to PD1 only group

B)

	%TGI
ZEN-3694-25 mg/kg	29.5
ZEN-3694-50 mg/kg	39.2
PD-1 5 mg/kg	54.6
ZEN-3694 25mg/kg+PD-1	74.7
ZEN-3694 50mg/kg+PD-1	80.6

Above additive

%TGI = Tumor Growth Inhibition

Figure 6. ZEN-3694 shows synergistic anti-tumor activity with anti-PD1 in the MC-38 model. A) MC-38 tumors were injected subcutaneously in C57Bl/6 mice. Once the tumors reached approximately 130 mm**³**, mice were dosed daily with ZEN-3694 given orally, or once every 3 or 4 days intraperitoneally with anti-PD1 (BioXcell). Mice were sacrificed for final tumor weights after 25 days of dosing. B) Final % TGI relative to vehicle.

ZEN-3694 has the potential to increase the anti-tumor immune response

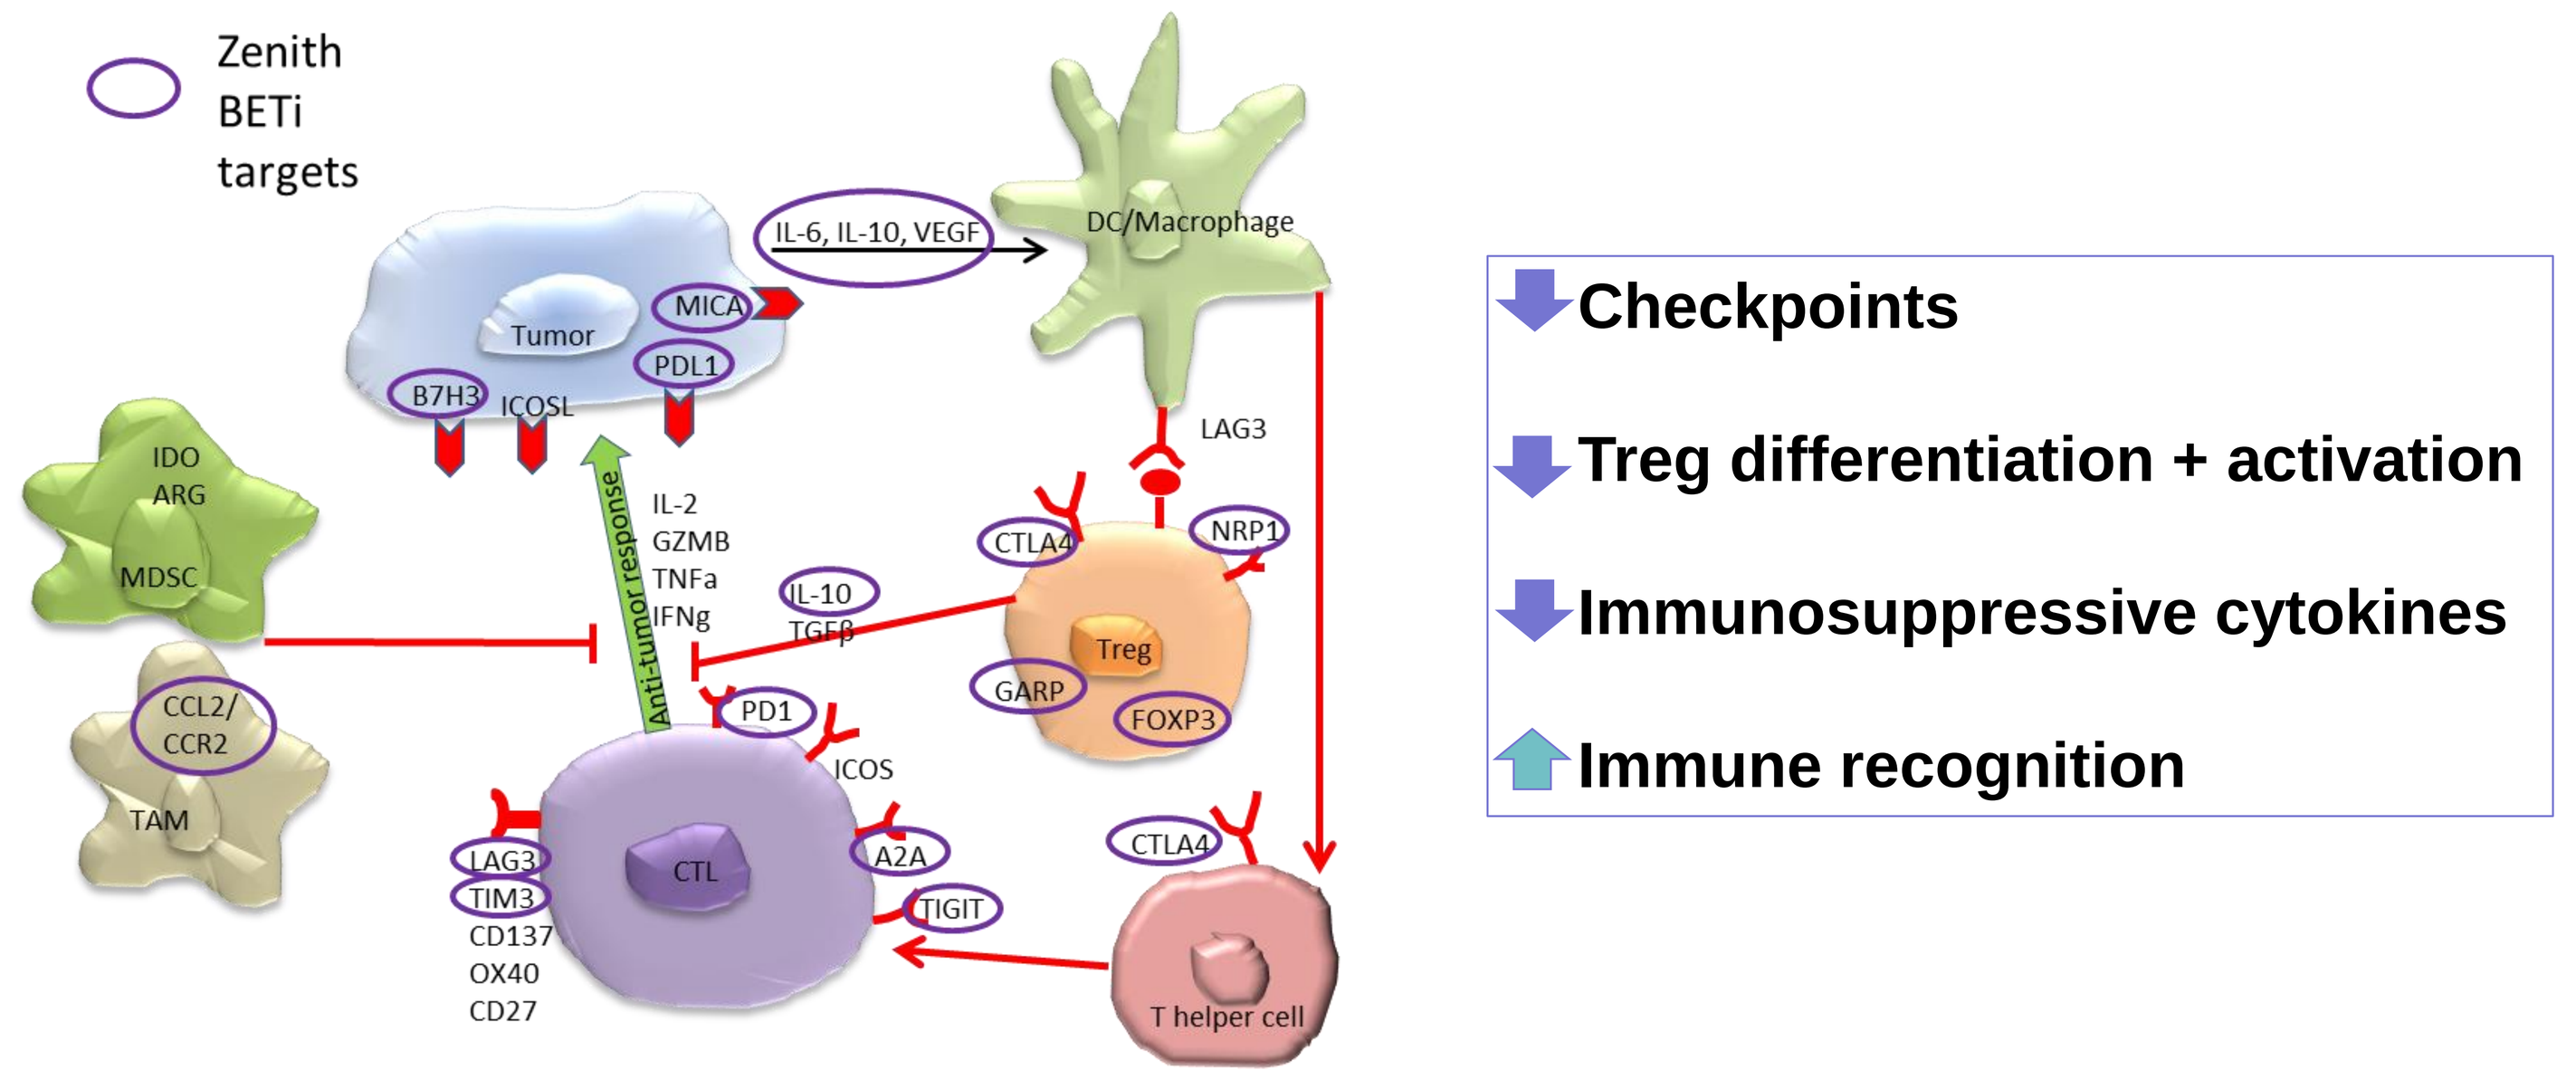
Zenith BETi targets
Tumor
MICA
PDL1
B7H3
ICOSL
IL-6, IL-10, VEGF
DC/Macrophage
IDO
ARG
MDSC
CCL2/ CCR2
TAM
Anti-tumor response
IL-2
GZMB
TNFa
IFNg
IL-10
TGFβ
LAG3
CTLA4
NRP1
Treg
GARP
FOXP3
PD1
ICOS
CTL
LAG3
TIM3
CD137
OX40
CD27
A2A
TIGIT
CTLA4
T helper cell
Checkpoints
Treg differentiation + activation
Immunosuppressive cytokines
Immune recognition

Figure 7. ZEN-3694 targets the expression of tumor and immune cell factors to increase the anti-tumor immune response. BETi have the potential to prime the tumor for increase immune recognition and killing, by inhibiting the checkpoints B7H3 and PD-L1, and increasing tumor antigen MICA expression. They also target suppressive cytokines such as IL-6, IL-10, and VegF. Furthermore, BETi target factors involved in the suppressive cell response, and ,most importantly, target multiple immune checkpoint receptors, suggesting that they may combine well with PD1 and other checkpoint antibodies

Summary

- **ZEN-3694 inhibits Treg cell differentiation and suppressive function.**
- **ZEN-3694 modulates a unique set of checkpoint receptors, cytokines and antigens on immune and tumor cells which were not targeted by the epigenetic modifier entinostat**
- **ZEN-3694 induces the tumor antigen MICA, and increases calreticulin cell surface expression, a marker of immunogenic cell death**
- **ZEN-3694 inhibits the mRNA expression of tumor PD-L1 and VegF, as well as the myeloid and macrophage suppressive cell markers CCR2, Cd11b, and CCL2, which could potentially stimulate the anti-tumor immune response**
- **ZEN-3694 increases the number of activated T cells in the tumor and draining lymph node**
- **ZEN-3694 synergizes with anti PD1 antibody to inhibit tumor growth in the MC-38 syngeneic tumor model**
- **ZEN-3694 has the potential to epigenetically prime the tumor to increase the frequency and duration of PD1 response, with a mechanism distinct from HDAC inhibitors**

References

1. Loven et al. (2013) Selective Inhibition of Tumor Oncogenes by Disruption of Super-Enhancers. *Cell* 153, 320–334
2. Hnisz et al. (2013) Super-Enhancers in the Control of Cell Identity and Disease. *Cell* 155, 1–14
3. Zou et al. (2014) Brd4 maintains Constitutively Active NF-kB in Cancer Cells by Binding to Acetylated RelA . *Oncogene* 33, 2395-404
4. Shi et al (2014) Disrupting the Interaction of BRD4 with Diacetylated Twist Suppresses Tumorigenesis in Basal-like Breast Cancer. *Cancer Cell* 25, 210–225